Characterization and Enumeration of Multiple Circulating Tumor Cell Phenotypes Using Two Distinct Platforms Establishes Presence of Epithelial-Mesenchymal Transition CTCs in Patients

Lihua Wang[1], Sonny Khin[1], Francis Owusu[1], Ralph E. Parchment[1], Alice Chen[2], Shivaani Kummar[2], James H. Doroshow[2], and Robert J. Kinders[1]

[1]Laboratory of Human Toxicology and Pharmacology, Applied/Developmental Directorate, Leidos Biomedical Research, Inc., Frederick National Laboratory for Cancer Research, Frederick, Maryland 21702; and [2]Division of Cancer Treatment and Diagnosis, National Cancer Institute, Bethesda, Maryland 20892

Introduction

CTCs are one form of liquid biopsy for measuring the pharmacodynamic effects of targeted anticancer drugs in clinical trials. In this poster, we present evidence that there are diverse phenotypes of CTCs that can be classified on the basis of established tumor and tissue markers, such as epithelial of tumor origin, sarcomas sourced, or epithelial-mesenchymal transitional CTCs, which include epithelial, endothelial and mesenchymal CTCs.

The current 4-color CellSearch method is dependent on expression of EpCAM, resulting in failure of CTC isolation from ~60% of patients with advanced, disseminated cancers, and cannot be applied to the collection of CTCs generated by sarcomas or lymphomas. The portion of the evaluable population is approximately 30% for all trials, and the CTC biomarker statistical evaluation is limited by the total number of CTCs collected from each tube of blood. Another limitation of the CellSearch instrument is that the number of imaging channels available precludes simultaneous positive identification of cancer cells using validated tumor markers and measurement of pharmacodynamic biomarkers.

To overcome these limitations, we have been applying the ApoStream platform (DEP-FFF separation) with fluorescence imaging and using the knowledge gained from that testing, we have recently added the 5-channel CellSearch system (Mab- coated ferrofluid) adding a fifth filter, PerCP, for specimen analysis.

A "home brew" CTC kit was developed that captures circulating cells that are either EpCAM or CD146 marker positive, and it was capable of identifying high numbers of EMT+ tumor marker+ double positive cells in patient specimens. Use of both CD146 and EpCAM capture per specimen, with the addition of vimentin and the tumor markers, significantly increased the number of captured CTCs that could be classified with the 5-channel CellSearch.

This approach solves a major limitation of using CTCs to monitor pharmacodynamic response by enumerating statistically significant cell numbers from patients with solid tumors of diverse histologies, using a smaller blood specimen.

Translational Relevance

- The application of CTCs in clinical studies is currently limited by:
 - Greater than 50% of patient CTCs not being EpCAM positive
 - Inability to move back and forth between bridge species because of low cell numbers and antibody cross-reactivity requirements
 - Difficulties in using isolated CTCs for downstream analysis (requires viable cells for expansion and increased purity)
 - Inability to isolate important EpCAM negative subpopulations, specifically EMT CTCs
 - Requirement for high blood volumes
 - Relatively low cell number collection making statistical analysis challenging
- A new technology (ApoStream) addresses these limitations and provides evidence of broad phenotypic plasticity among CTCs from individual carcinoma and sarcoma patients
- New generation CellSearch 5-color system confirms ApoStream results and allows for the use of fixed cells

Methods

Patients and Sample Collection

- All enrolled patients and healthy subjects gave informed consent for study inclusion and were enrolled using institutional review board–approved protocols.
- Blood was drawn from metastatic prostate, breast, or other cancer patients at DCTD/NCI and other clinical centers (USC, UCD, COH, DFCI).
- Blood (~10 mL) was collected into CellSave® tubes (Veridex) and processed within 96 hours.
- 4-mL heparinized blood collections were used for ApoStream analysis and processed the day of collection

CTC Enumeration and Identification in Patient Blood

- For CellSearch, a blood sample (2 to 7.5 mL) was mixed with 6.5 to 12 mL sample buffer, and was centrifuged and processed using the CellSearch platform (Janssen) and CXC kit or CEC kit.
- For ApoStream, a 4-mL blood specimen was Ficoll separated and then processed through the flow chamber, and was collected and spotted onto a Marienfeld slide for image analysis
- Images captured by the 5-color system in CellTracks® Analyzer II contain objects fulfilling predetermined criteria and are automatically presented in gallery format. Final classification of cells is done independently by two operators.
- Cells are classified as CTCs when morphologic features and staining patterns are consistent with that of epithelial cells (CK-PE positive, DAPI positive, CD45-APC negative, and tumor marker positive) or mesenchymal cells (Vimentin positive, DAPI positive, CD45-APC negative, and tumor marker positive).
- CTCs must have a minimum size of at least 4 µm, but present with a large heterogeneity in both CTC size and morphology.
- γH2AX-positive CTCs present with nuclear staining in the PerCP channel.

References

1. Lihua Wang, Thomas D. Pfister, Ralph E. Parchment, Shivaani Kummar, Larry Rubinstein, Yvonne A. Evrard, Martin E. Gutierrez, Anthony J. Murgo, Joseph E. Tomaszewski, James H. Doroshow, and Robert J. Kinders. Monitoring Drug-Induced γH2AX as a Pharmacodynamic marker in Individual circulating tumor cells. Clinical Cancer Research. 2010, 16:1073-1084.
2. Priya Balasubramanian, Lihua Wang, Scott M. Lawrence, Tony Navas, Shivaani Kummar, Melinda Hollingshead, Francis Owusu, Ralph E. Parchment, Joseph E. Tomaszewski, James H. Doroshow, Robert J. Kinders. Isolation and characterization of circulating tumor cells (CTCs) from peripheral blood specimens of patients with advanced solid tumor malignancies (using ApoStream™ instrumentation). 2014 AACR Annual Meeting. Abstract and poster. April, San Diego, CA.

Acknowledgements

This project has been funded in whole or in part with federal funds from the National Cancer Institute, National Institutes of Health, under Contract No. HHSN261200800001E.

This research was supported in part by the Intramural Research Program of the NIH and the Developmental Therapeutics Program in the Division of Cancer Treatment and Diagnosis of the National Cancer Institute.

The CTC specimens for this study were provided by DCTD/NCI and other clinical centers at City of Hope Medical center, USC Norris Comprehensive Cancer Center, UC-Davis Comprehensive Cancer Center, Dana Farber Cancer Institute, and Yale Cancer Center.

The CellSearch 5-Channel Platform was provided by Janssen Diagnostics, and PADIS/FNLCR serves as a beta testing site for this device.

Results

CTC and Biomarker-positive CTC Enumeration Comparison

	Before Installation		After Installation	
Patient samples analyzed using CTC kit	CTC #	γH2AX-positive CTC #	CTC #	γH2AX-positive CTC #
5-#1 (DFCI 056 C1D9)	45	9	45	9
5-#2 (DFCI 056 C1D10)	22	0	22	0
Patient samples analyzed using CEC kit	CEC #	γH2AX-positive CEC #	CTC #	γH2AX-positive CEC #
5-#3 (DFCI 058 C1D1)	57	12	57	12
5-#4 (JHU 32 C1D12)	24	6	24	6
Spike samples analyzed using CTC kit	CTC #	γH2AX-positive CTC #	CTC #	γH2AX-positive CTC #
5-#5 (HT29 cells were treated with 1 µM Topotecan for 2 hrs)	480	52	480	52
5-#6 (MCF7 cells were treated with 1 µM Topotecan for 2 hrs)	506	58	506	58

Setup of CellSearch CTC Assays with Home Brew CTC Kit Used for 5-Color System Versus the 4-Color System

	5-Color system		4-Color system
Ferrofluid capture (Enrichment)	EpCAM + CD146		EpCAM
Images presented in FITC channel (Positive selection & cell type identification)	CK-FITC DAPI	VIMENTIN-AF-488 DAPI	CK-PE DAPI
Leukocyte maker (Negative selection)	CD45-APC		CD45-APC
Tumor markers (CTC characterization)	CEA-PE+ MUC1-PE S100-PE PSA-PE+PSMA-PE		No
PD marker (PD marker identification)	γH2AX -PerCP		γH2AX -FITC

CTC Count in Healthy Donor Blood Measured by the 5-Color System with Two Types of Modified CEC Kits

	5-Color (Home brew kit)			
	CEC-CK kit (7.5 mL)		CEC-VIMENTIN kit (2 mL)	
Sample ID	Rare Cells (CK+, CEA/MUC1-, CD45-)	Epithelial CTCs (CK+, CEA/MUC1+, CD45-)	Rare Cells (VIM+, CEA/MUC1-, CD45-)	Mesenchymal CTCs (VIM+, CEA/MUC1+, CD45-)
RDP0530	3	0	0	0
RDP0459	1	0	2	0
RDP0299	0	0	0	0

CTC Count in Healthy Donor Blood Measured by the 5-Color System with Two Types of Modified CXC Kits

	5-Color (Home brew kit)			
	CXC-CK kit (7.5 mL)		CXC-VIMENTIN kit (2 mL)	
Sample ID	Rare Cells (CK+, CEA/MUC1-, CD45-)	Epithelial CTCs (CK+, CEA/MUC1+, CD45-)	Rare Cells (VIM+, CEA/MUC1-, CD45-)	Mesenchymal CTCs (VIM+, CEA/MUC1+, CD45-)
RDP0763	0	0	3	1
RDP0027	1	0	0	0
RDP0044	0	0	1	0

ApoStream Workflow

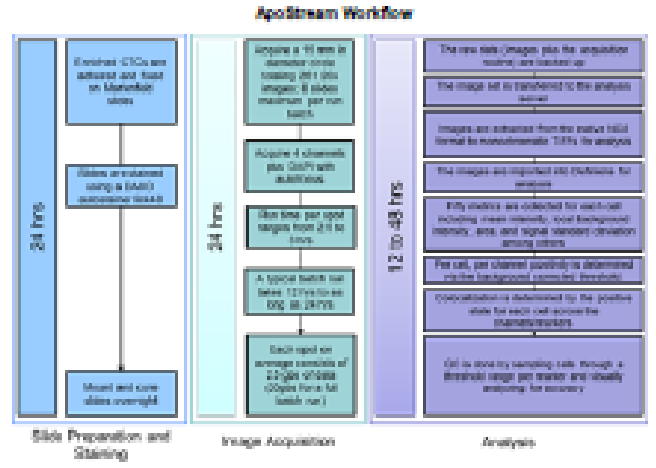

24 hrs
24 hrs
12 to 48 hrs
Slide Preparation and Staining
Image Acquisition
Analysis

Various Types of CTCs Detected by the 5-Color System Using Our Home Brew CTC Kits

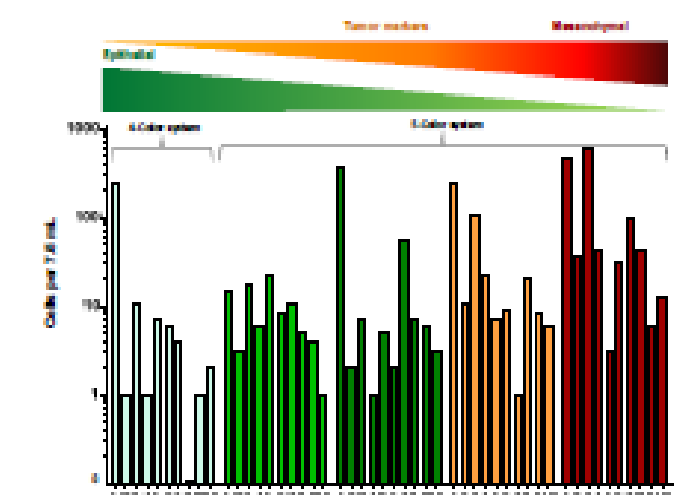

Tumor markers
Mesenchymal
Epithelial
4-Color system
5-Color system
Cells per 7.5 mL

EpCAM	+ (4-Color)	5-Color system			
EpCAM/CD146	n/a	+	+	+	+
CK 8, 18, 19	+	-	+	+	-
CEA/MUC1	n/a	+	+	+	+
Vimentin	n/a	-	-	+	+
DAPI	+	+	+	+	+
CD45	-	-	-	-	-

CD45-/Tumor Marker+ Circulating Cells at Baseline, Developmental Therapeutics Clinic

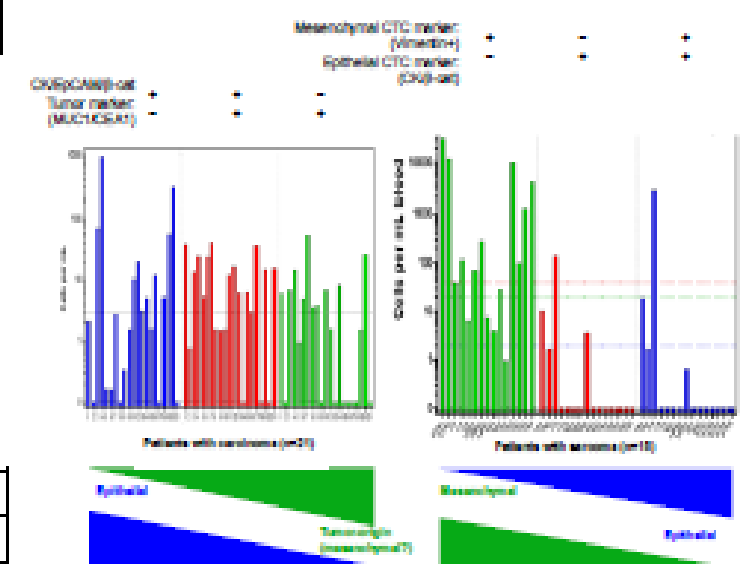

Mesenchymal CTC marker: (Vimentin+)
Epithelial CTC marker: (CK8-ert)
CK/EpCAM/β-cat
Tumor marker: (MUC1/CEA1)
Cells per mL blood
Patients with carcinoma (n=21)
Patients with sarcoma (n=18)
Epithelial
Tumor origin (mesenchymal?)
Mesenchymal
Epithelial

- Heterogeneity in CTCs isolated in an EpCAM-independent manner
- Individual patients show multiple phenotypes
- EMT in carcinomas (CEA/MUC1+); MET in sarcomas (TLE/ASPL-1/MUC1+)
- Sarcoma phenotyping is complex, requiring a large variety of tumor markers

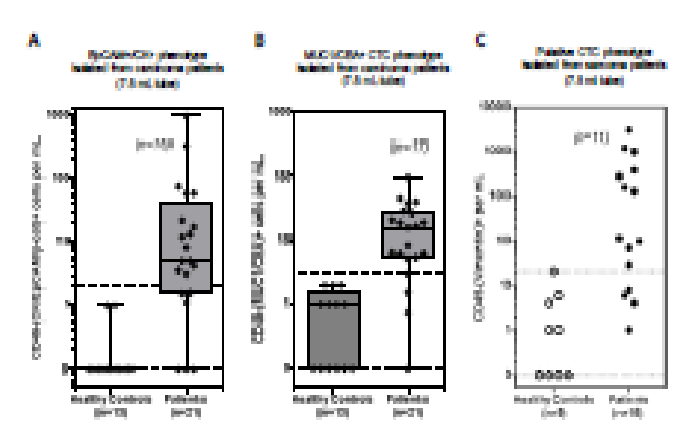

A
B
C
Healthy Controls
Patients

- ApoStream is able to isolate CTCs from patients with a range of carcinomas and sarcomas
- Increased numbers of (tumor-marker positive) CTCs from carcinomas as compared to EpCAM+ capture

Triple-Negative Breast Cancer Cell Line MDA-MB-231 and Endothelial Cell Line HUVEC Were Spiked into Healthy Donor Blood and Processed by the CEC Kit Using the CellSearch System

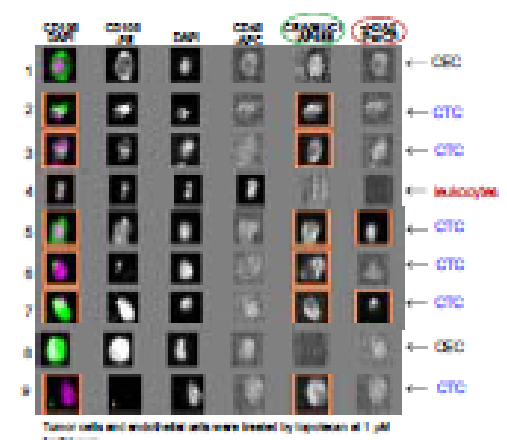

DAPI
CEC
CTC
CTC
leukocytes
CTC
CTC
CTC
CEC
CTC

Tumor cells and endothelial cells were treated by topotecan at 1 µM for 2 hours

CTC Images from Post-treatment Specimen of a Patient by the 5-Color CellSearch System

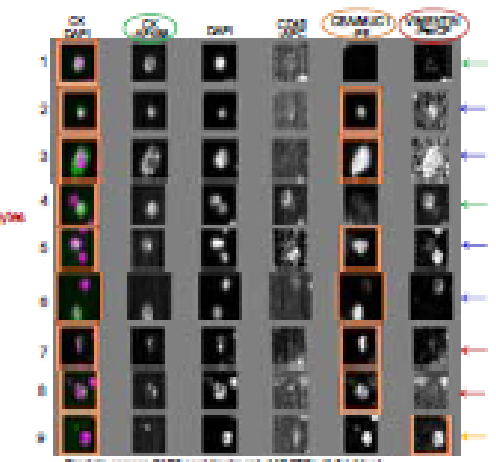

DAPI

Prostate cancer, C1D8 post-treatment, 168 CTCs / 1.5 L blood

CTC Images from Pre-treatment Specimen of a Patient Detected by the 5-Color CellSearch System Using the Modified CXC Kit

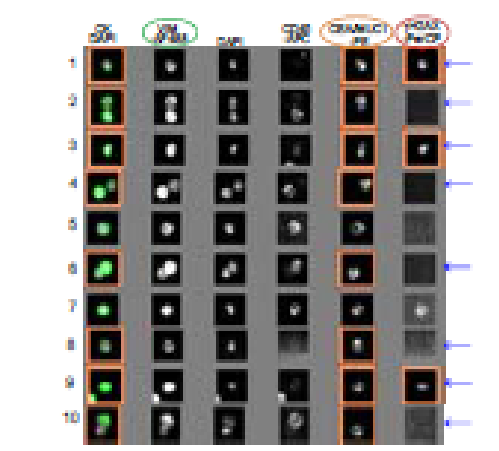

DAPI

Uterine cancer, C1D15, post-treatment, 103 CTCs / 2 mL blood

Leiomyosarcoma Cell Line SK-LMS-1 and Endothelial Cell Line HUVEC Were Spiked into Healthy Donor Blood and Analyzed Using the CEC Kit by the 5-Color System

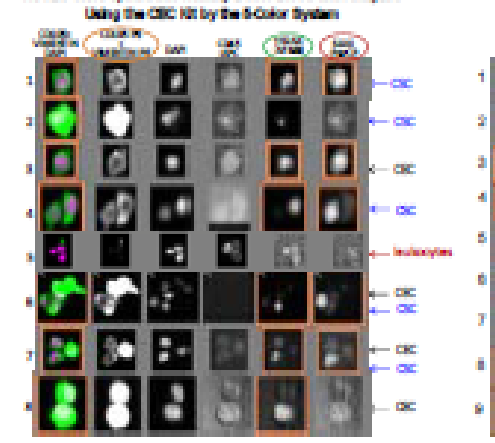

DAPI
CEC
CEC
CEC
CEC
leukocytes
CEC
CEC
CEC
CEC
CEC

Tumor cells and endothelial cells were treated by topotecan at 1 µM for 2 hours

CTC Images from Post-treatment Specimen of a Patient by the 5-Color CellSearch System

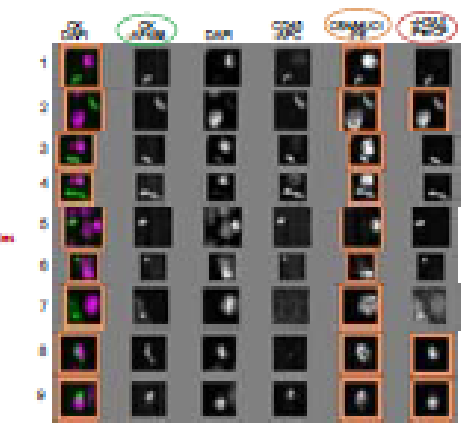

DAPI

Triple-negative breast cancer, C1D1 pre-treatment, 181 CTCs / 1.7 mL blood

Comparison of CTC Count Measured by Different Systems

Specimen ID (CTEP #9484)	Diagnosis	4-color (CTC kit) (7.5 mL blood)	5-color (CXC kit)	
			Epithelial CTC (CK-, DAPI+, CEA/MUC1+, VIM-, CD45-)	Mesenchymal CTC (CK-, DAPI+, CEA/MUC1+, VIM+, CD45-)
DFCI 060 C1D1	Uterine Ca	7	[illegible] mL, 11 (1.8 mL blood)	[illegible] mL, 20 (1.8 mL blood)
DFCI 060 C1D8	Uterine Ca	14	[illegible] mL, 28 (2.0 mL blood)	[illegible] mL, 108 (2.0 mL blood)
DFCI 060 C1D8	Uterine Ca	8	[illegible] mL, 16 (2.0 mL blood)	[illegible] mL, 60 (2.0 mL blood)
DFCI 060 C1D15	Uterine Ca	1	[illegible] mL, 8 (2.0 mL blood)	[illegible] mL, 117 (2.0 mL blood)
DFCI 084 C1D1	Triple negative breast Ca	2	[illegible] mL, 13 (1.8 mL blood)	[illegible] mL, 88 (1.8 mL blood)

CTC Characterization and PD Marker Assessment Using the 5-Color CellSearch System

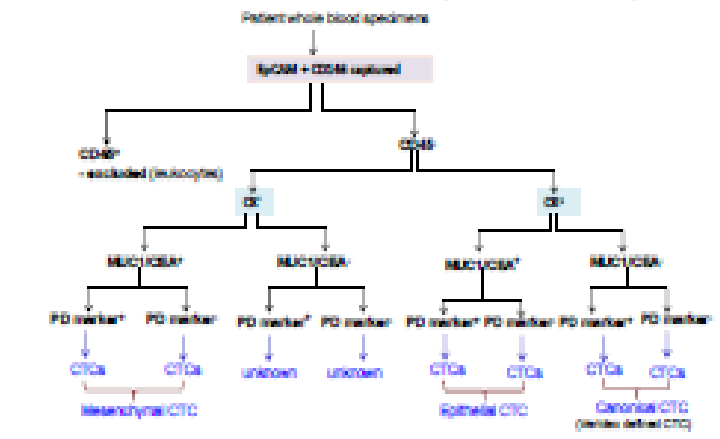

Patient whole blood specimens
EpCAM + CD146 captured
CD45+
- excluded (leukocytes)
CD45-
CK-
CK+
MUC1/CEA+
MUC1/CEA-
PD marker+
PD marker-
CTCs
unknown
Mesenchymal CTC
Epithelial CTC
Canonical CTC
(Veridex defined CTC)

CTC Characterization and PD Marker Assessment Using the 5-Color CellSearch System

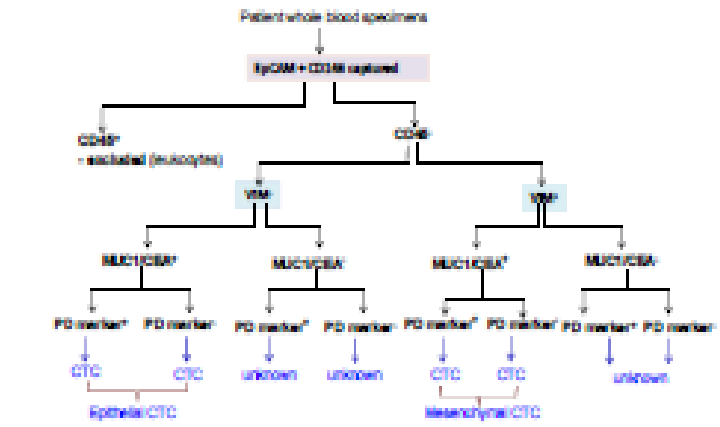

Patient whole blood specimens
EpCAM + CD146 captured
CD45+
- excluded (leukocytes)
CD45-
VIM-
VIM+
MUC1/CEA+
MUC1/CEA-
PD marker+
PD marker-
CTC
unknown
Epithelial CTC
Mesenchymal CTC

Summary and Conclusions

- Two platforms, ApoStream and the 5-color CellSearch system, have been applied for monitoring drug responses to new anticancer agents in early stage clinical trials by measuring drug effects in circulating tumor cells (CTCs).
- A "home brew" CTC kit was developed that captures circulating cells that are either EpCAM or CD146 marker positive, and it was capable of identifying high numbers of EMT+ tumor marker+ double-positive cells in patient specimens.
- There was statistical equivalency of events flagged as CTCs (CD45-/EpCAM+/CK+) in patient specimens analyzed using ApoStream or the 5-color CellSearch versus the 4-color CellSearch, and 100% concordance between the CellSearch platforms.
- Tumor marker specificity was established by analyzing normal donor blood, which varied from 0 to 2 cells per ml of blood depending on the marker set employed (only CD45-negative cells were assessed).
- Both platforms demonstrated the presence of high numbers of CTCs with the epithelial-mesenchymal transition (EMT) phenotype, and widely used tumor markers such as MUC1, CEA, TLE1, or proteins generated by recombination events such as ASPL-TFE3.
- The presence of large numbers of CTCs from carcinoma patients expressing the EMT phenotype may be related to the intensive prior treatment (>3 prior therapies, median) of our phase 1 population.

Frederick National Laboratory for Cancer Research

Confidential

leidos
Leidos Biomedical Research, Inc.